STX-003: Utilizing Programmable Genetic Circuits for Tumor-Specific IL-12 Expression and Reduced Toxicity with Systemically Delivered mRNA

Gautam N. Shenoy, Wen Allen Tseng, Emily A. Clinton, Ryan A. Patenaude, Wuhbet Abraham, Zhijun Su, Madhuri Dey, Justin H. Letendre, Rachel G. Eichman, Ashley G. Njiru, John T. Sexton, Emily Tjon, Dahyana Arias Escayola, Nandita Satish Menon, Isma Zarin, Breanna Burgett, Sarah O'Bryan, Mae Kingsbury, Aidan Coia, Akshay Mavani, Weiyu Zhao, Kevin Shi, Alexander C. Lemaire, Julian Quiñones, Ryan T. Sowell, Jaspreet S Khurana, Joseph Barberio, Prashant R. Nambiar, Jacob R. Becraft, Tasuku Kitada

Strand Therapeutics, Inc. 20 Overland St, Boston, MA 02215, USA

INTRODUCTION

Immunotherapies have revolutionized the treatment of solid tumors, which represent 90% of adult human cancers. Given the double-edged nature of immune-based interventions, there is increased emphasis on developing precision therapies that target tumors while having minimal effects on healthy tissues. IL-12, a potent immunostimulatory cytokine, is robustly effective in terms of anti-tumor activity in preclinical models. However, systemically delivered IL-12 is poorly tolerated potentially due to its off-target activity. To overcome this challenge, we developed an mRNA platform that utilizes programmable genetic circuits to regulate the expression of encoded protein based on specific molecular cues, enabling precise control of payload expression in the target tissues. Using this platform we developed STX-003, a self-replicating mRNA encoding IL-12 which bears programmable genetic circuitry that limits expression of payload in the spleen, while preserving expression in tumors.

IN SILICO MODELING INFORMS CIRCUIT SELECTION

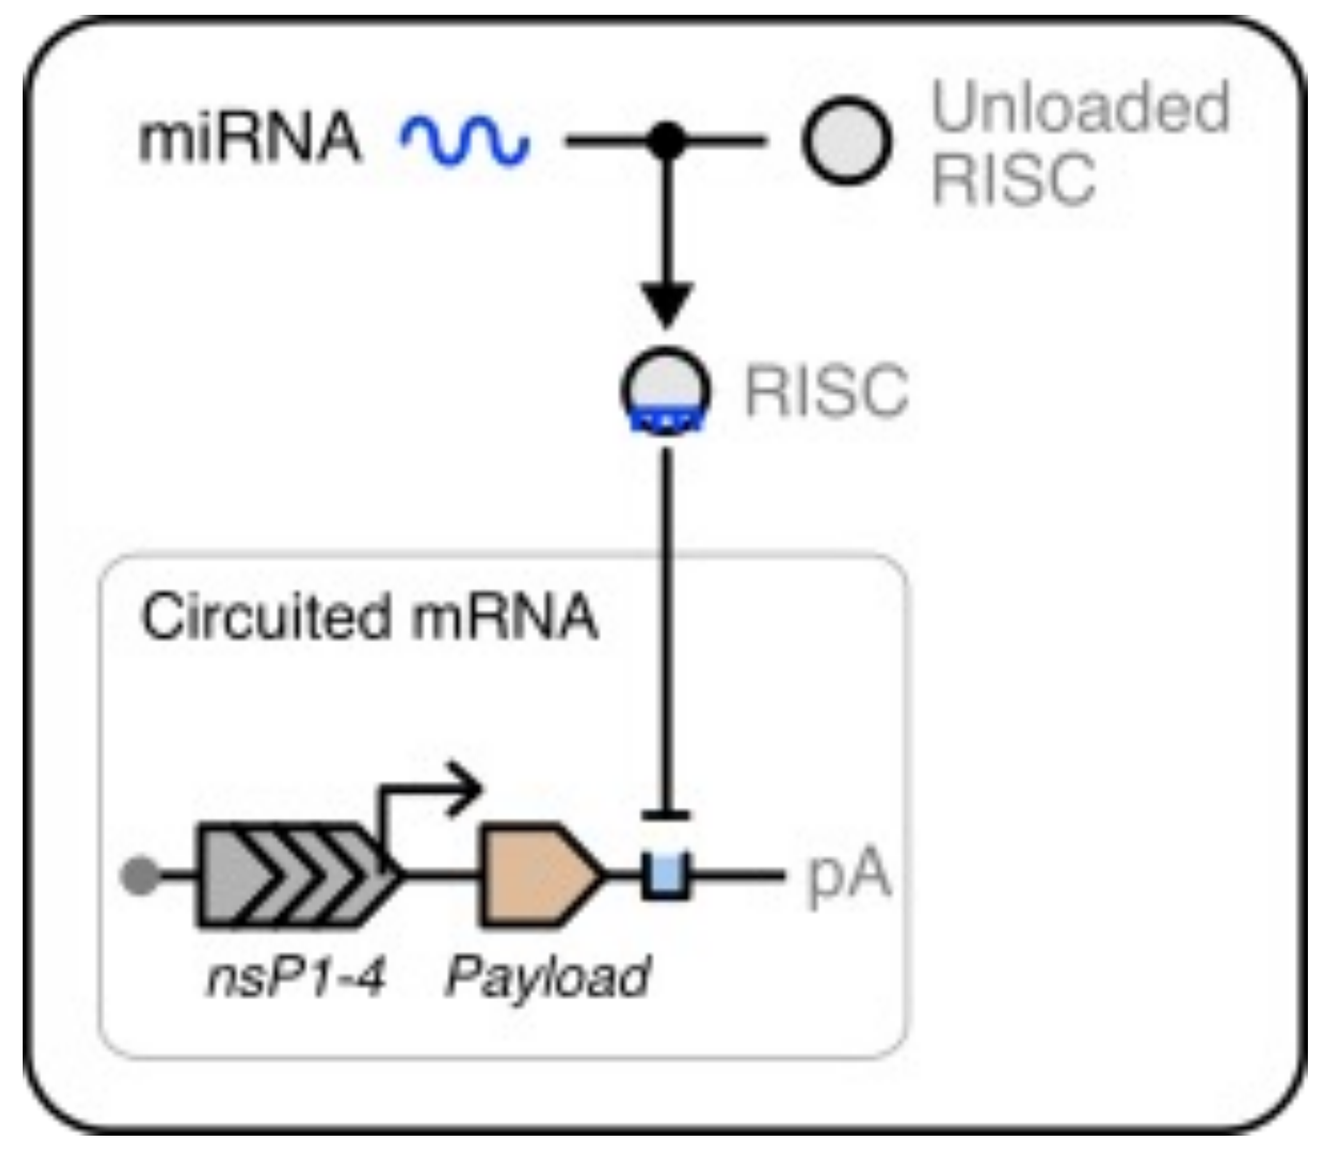
miRNA
Unloaded RISC
RISC
Circuited mRNA
pA
nsP1-4
Payload

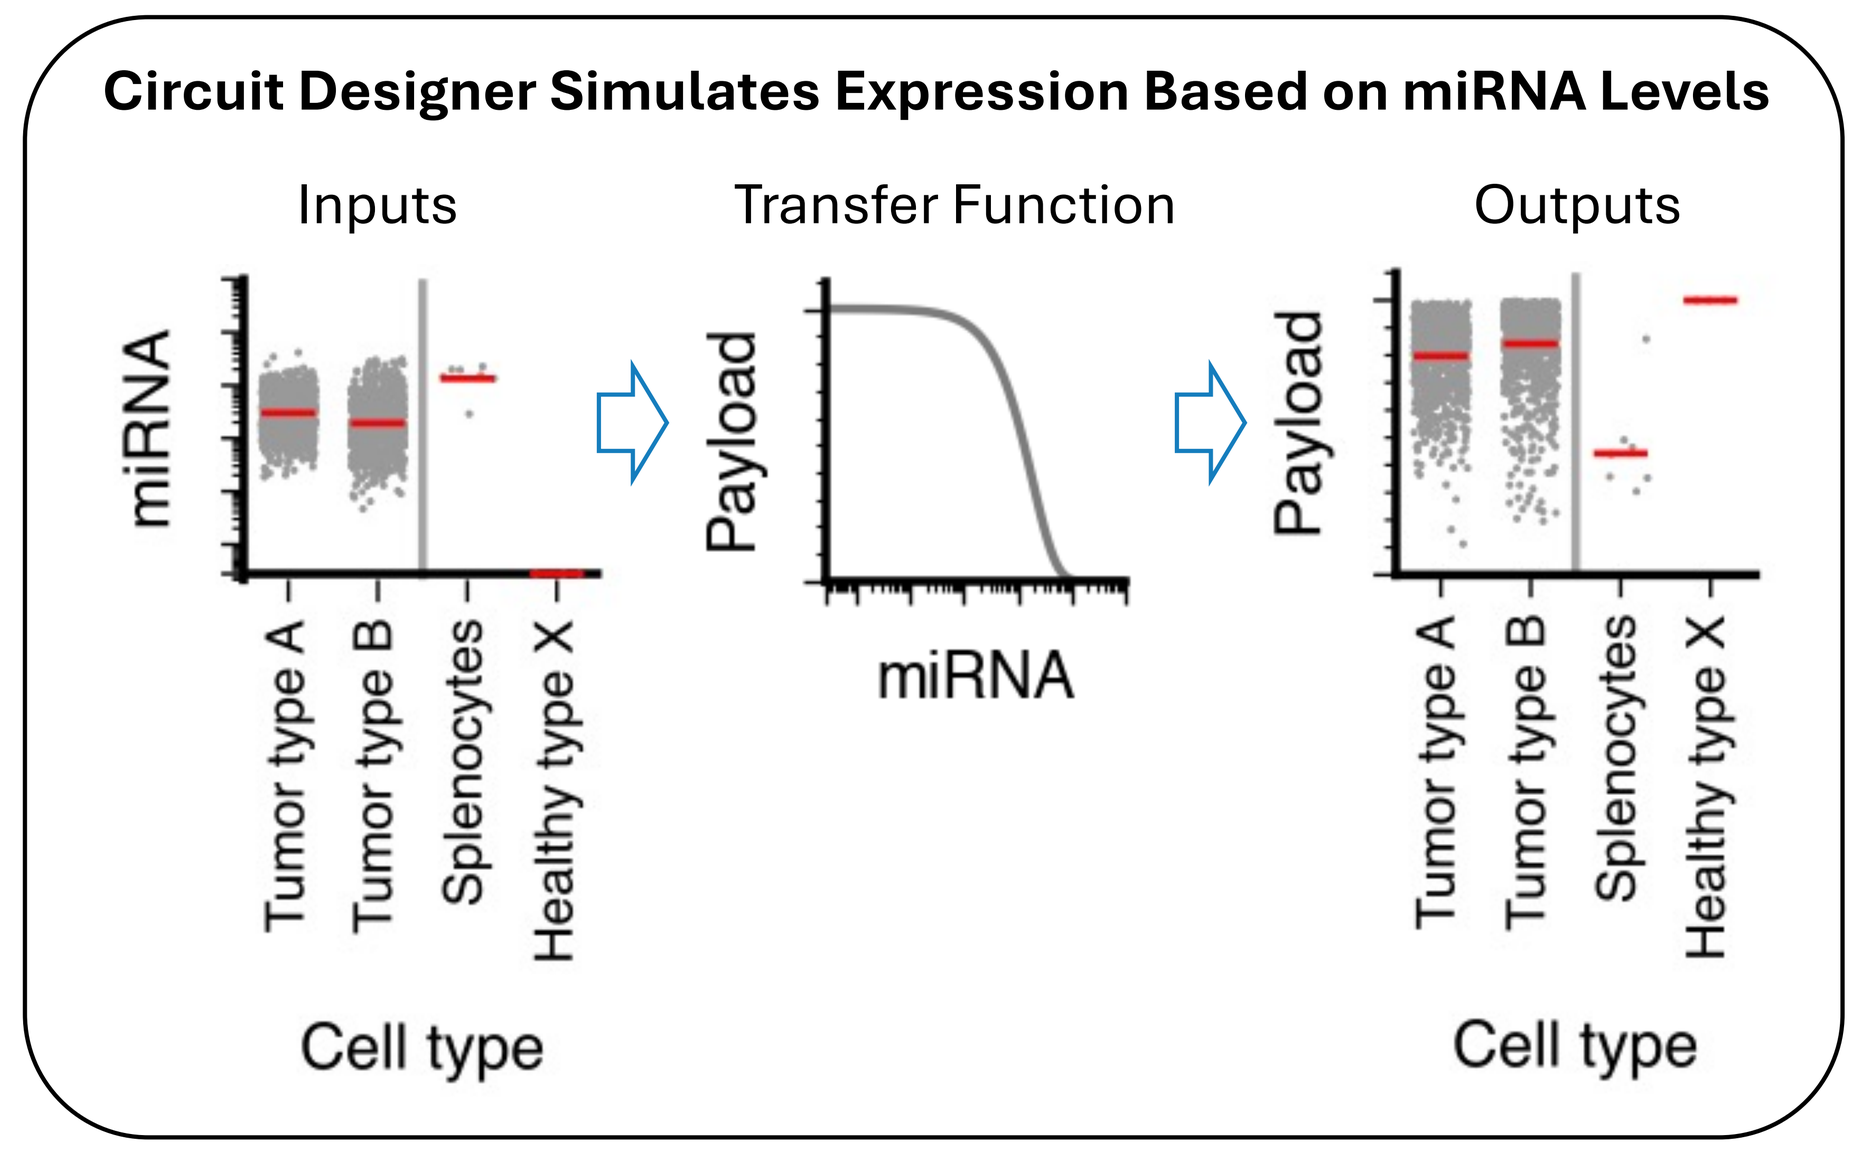
Circuit Designer Simulates Expression Based on miRNA Levels
Inputs
Transfer Function
Outputs
miRNA
Payload
Tumor type A
Tumor type B
Splenocytes
Healthy type X
Cell type

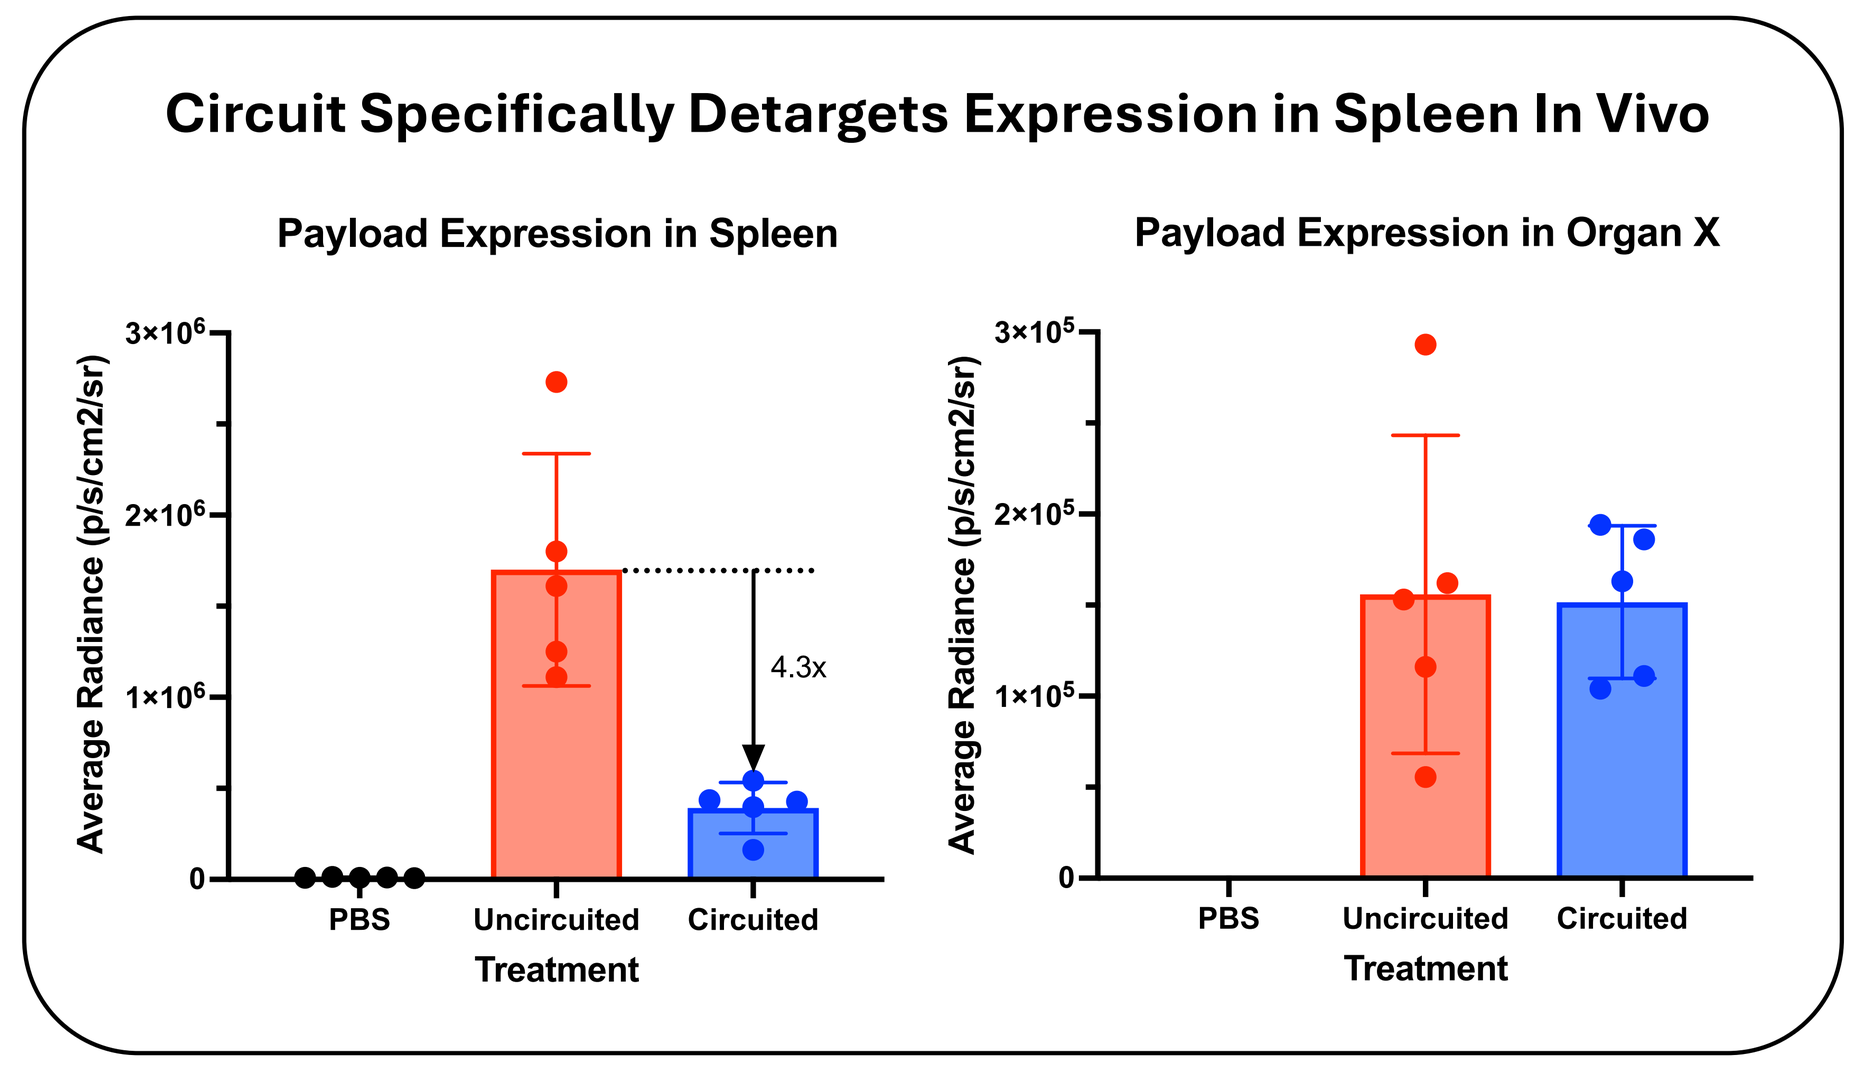
Circuit Specifically Detargets Expression in Spleen In Vivo
Payload Expression in Spleen
Payload Expression in Organ X
Average Radiance (p/s/cm2/sr)
4.3x
PBS
Uncircuited
Circuited
Treatment

- Unique miRNA signatures were identified using *in silico,* and high throughput *in vitro* screening.
- Candidate circuit was selected based on characteristics of detargeting the spleen.
- Circuited mRNA were formulated into LNPs and administered IV in mouse models to conduct proof-of-concept studies.

CIRCUIT IMPROVES TOLERABILITY IN NAÏVE MICE

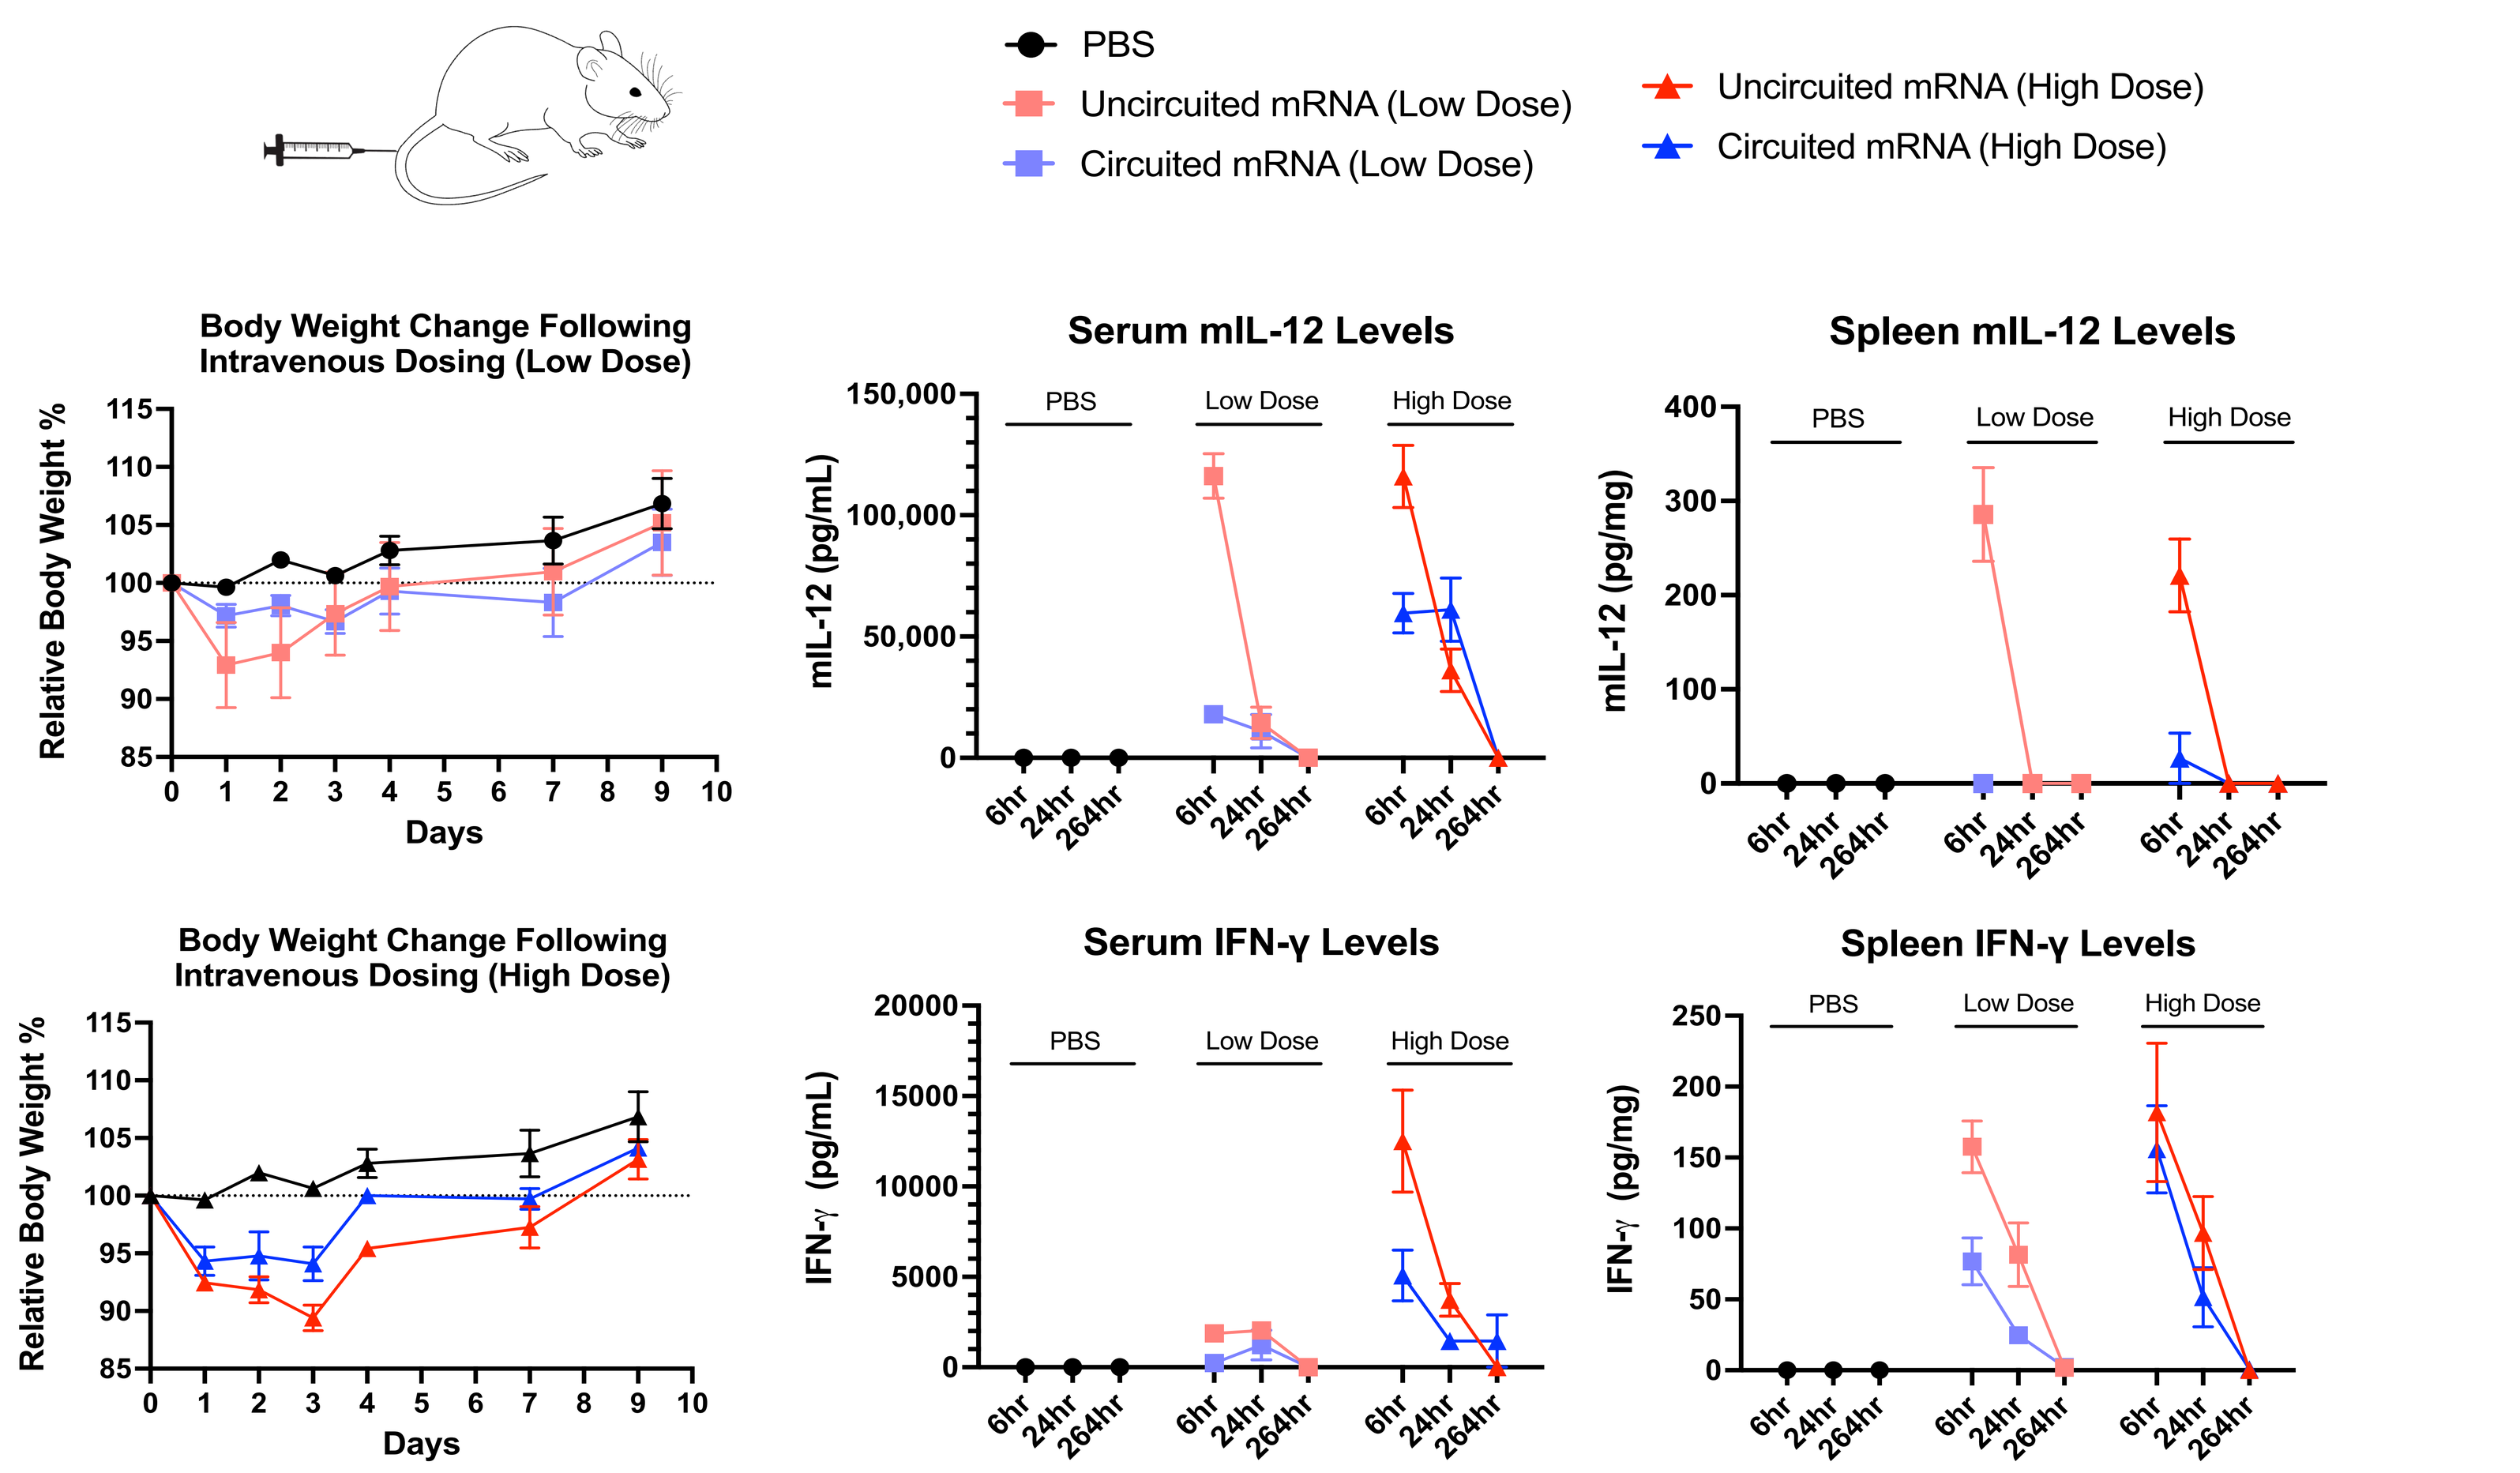
PBS
Uncircuited mRNA (Low Dose)
Circuited mRNA (Low Dose)
Uncircuited mRNA (High Dose)
Circuited mRNA (High Dose)
Body Weight Change Following Intravenous Dosing (Low Dose)
Body Weight Change Following Intravenous Dosing (High Dose)
Relative Body Weight %
Days
Serum mIL-12 Levels
Spleen mIL-12 Levels
Serum IFN-γ Levels
Spleen IFN-γ Levels
mIL-12 (pg/mL)
mIL-12 (pg/mg)
IFN-γ (pg/mL)
IFN-γ (pg/mg)
PBS
Low Dose
High Dose
6hr
24hr
264hr

- The presence of the circuit lowered test article-related body weight loss as well as recovery time in naïve C57BL6/J mice.
- This correlated with lower levels of splenic and circulating IL-12 and IFN-γ.

CIRCUIT REPRESSES PAYLOAD IN NON-HUMAN PRIMATES WHILE PRESERVING EXPRESSION IN CANCER CELLS

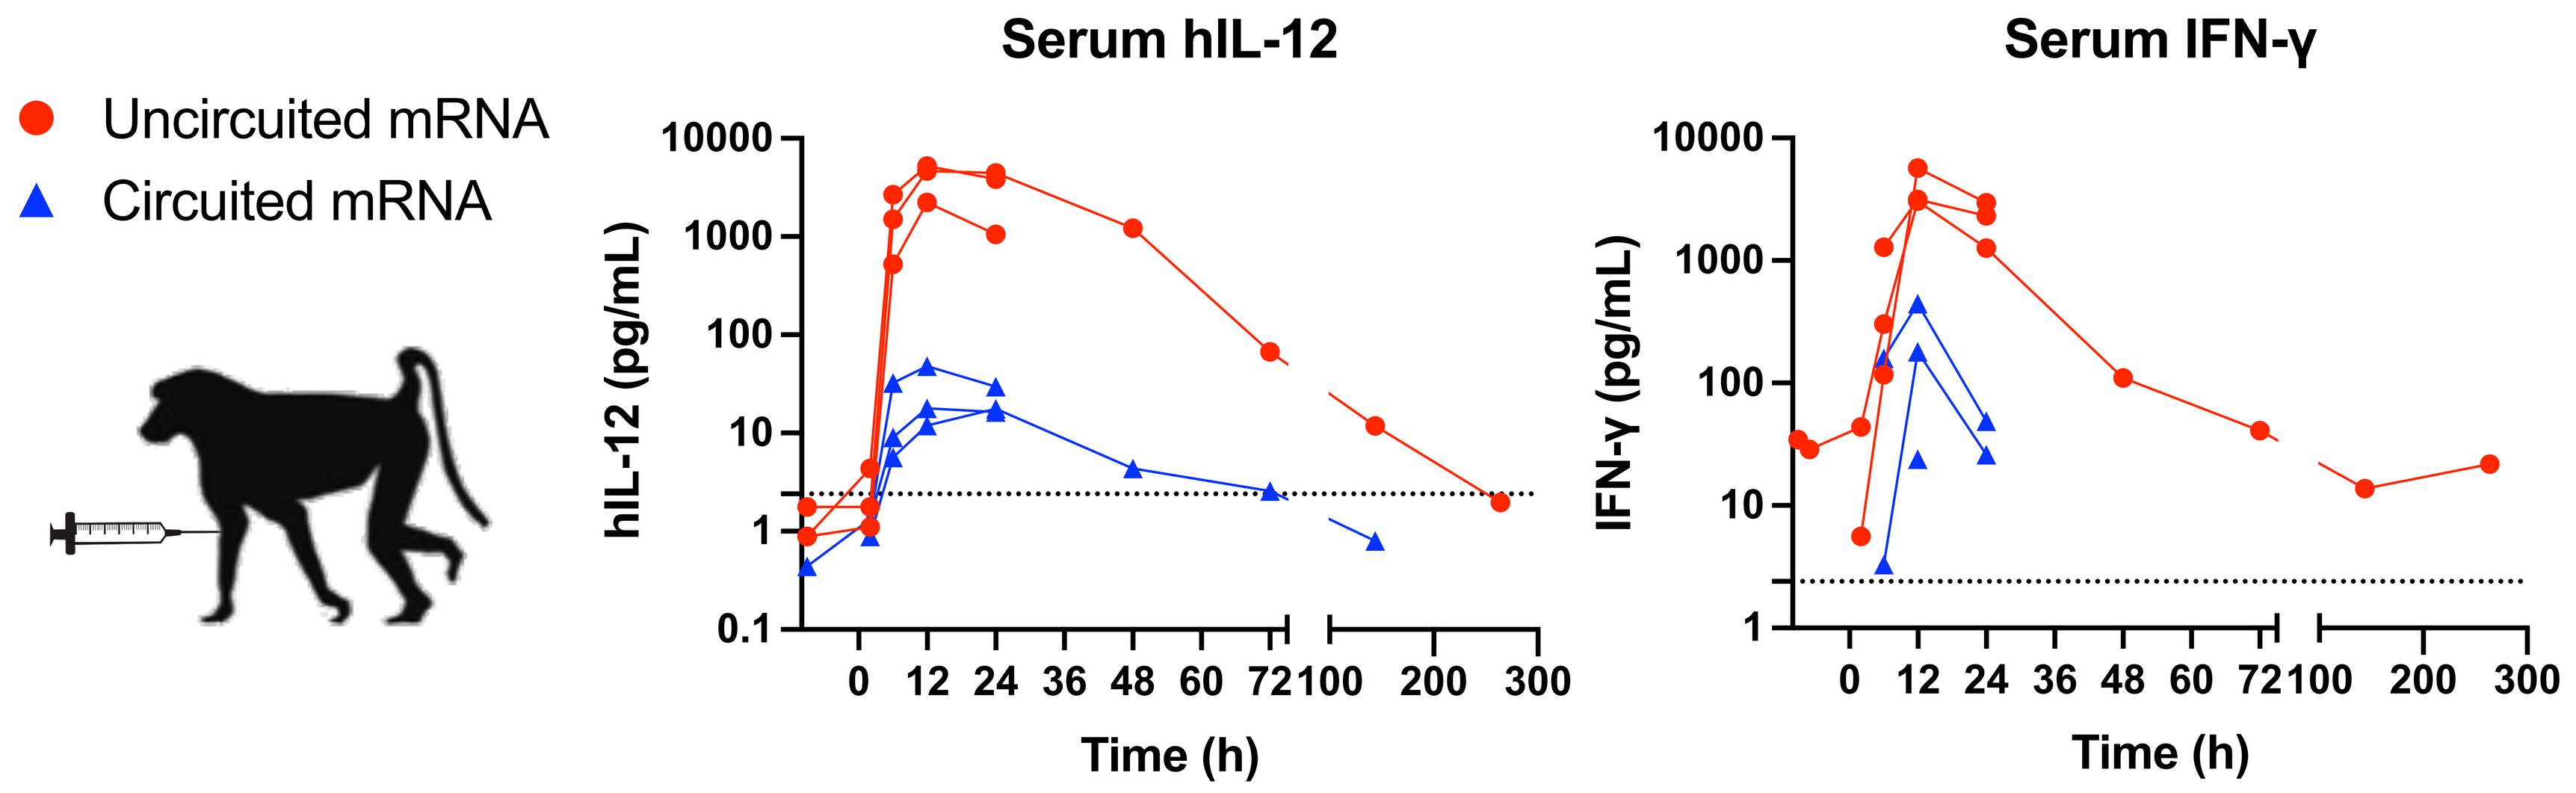
Uncircuited mRNA
Circuited mRNA
Serum hIL-12
Serum IFN-γ
hIL-12 (pg/mL)
IFN-γ (pg/mL)
Time (h)

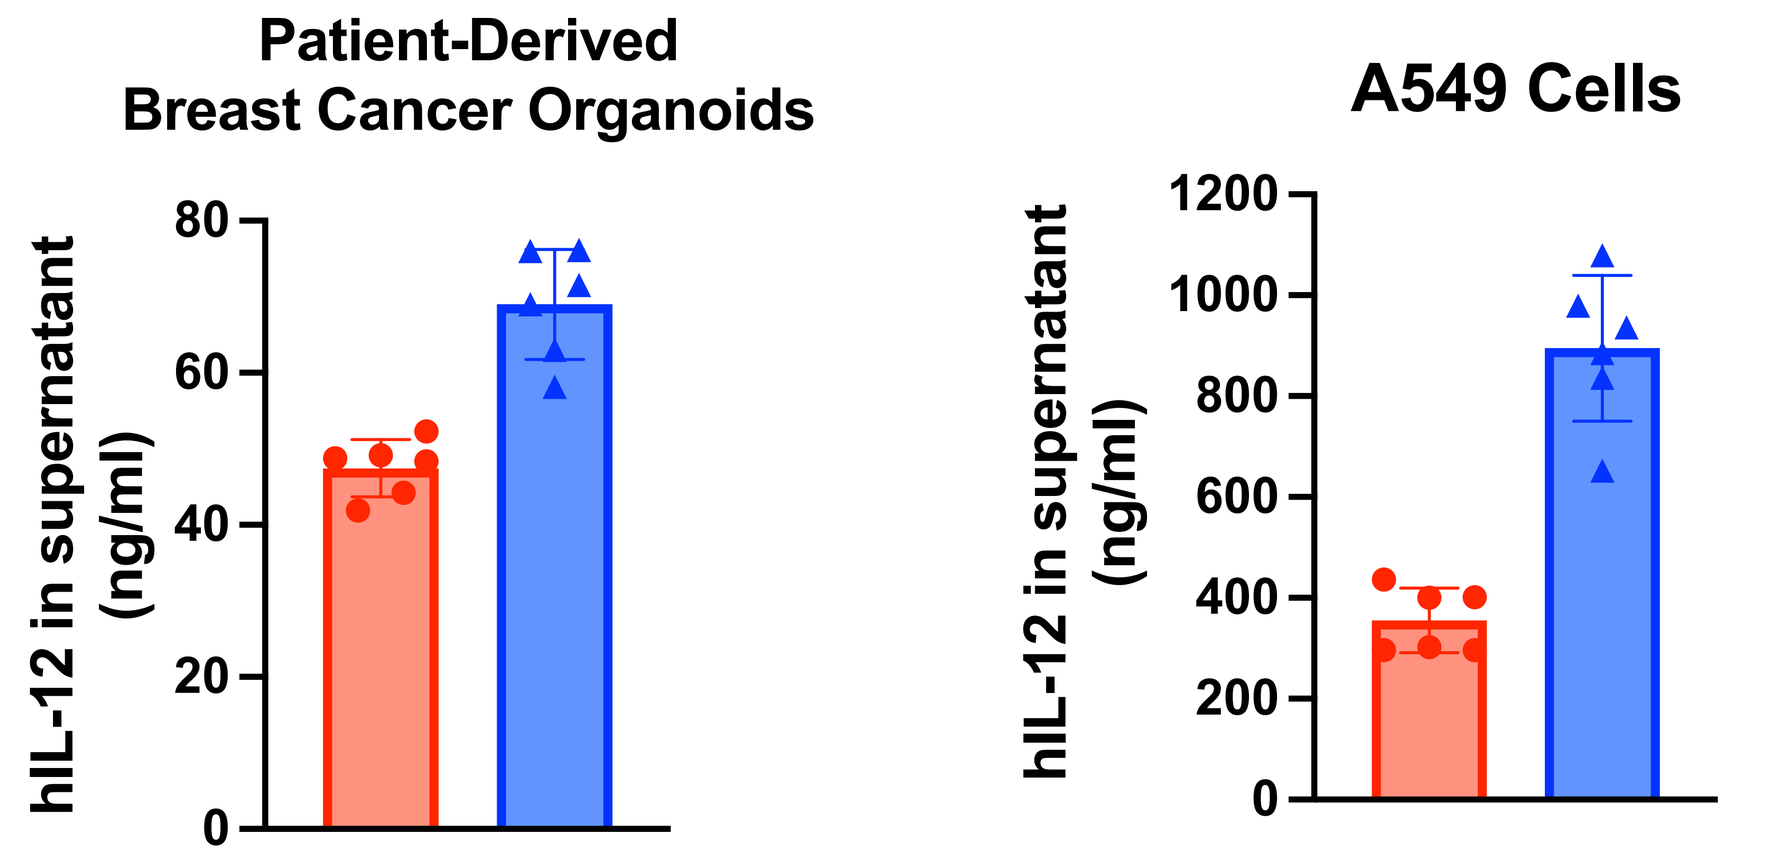
Patient-Derived Breast Cancer Organoids
A549 Cells
hIL-12 in supernatant (ng/ml)

- Cynomolgus monkeys were intravenously injected with mRNA-LNP.
- The presence of the circuit resulted in a ~99% reduction in circulating hIL-12 and ~95% reduction in circulating IFN-γ.
- Transfection of circuited and uncircuited mRNA into cancer organoids or A549 cancer cells yielded comparably robust expression of hIL-12 payload.

CIRCUIT DEMONSTRATES SPLENIC DETARGETING WHILE MAINTIANING EFFICACY IN B16-F10 ONCOLOGY MODEL

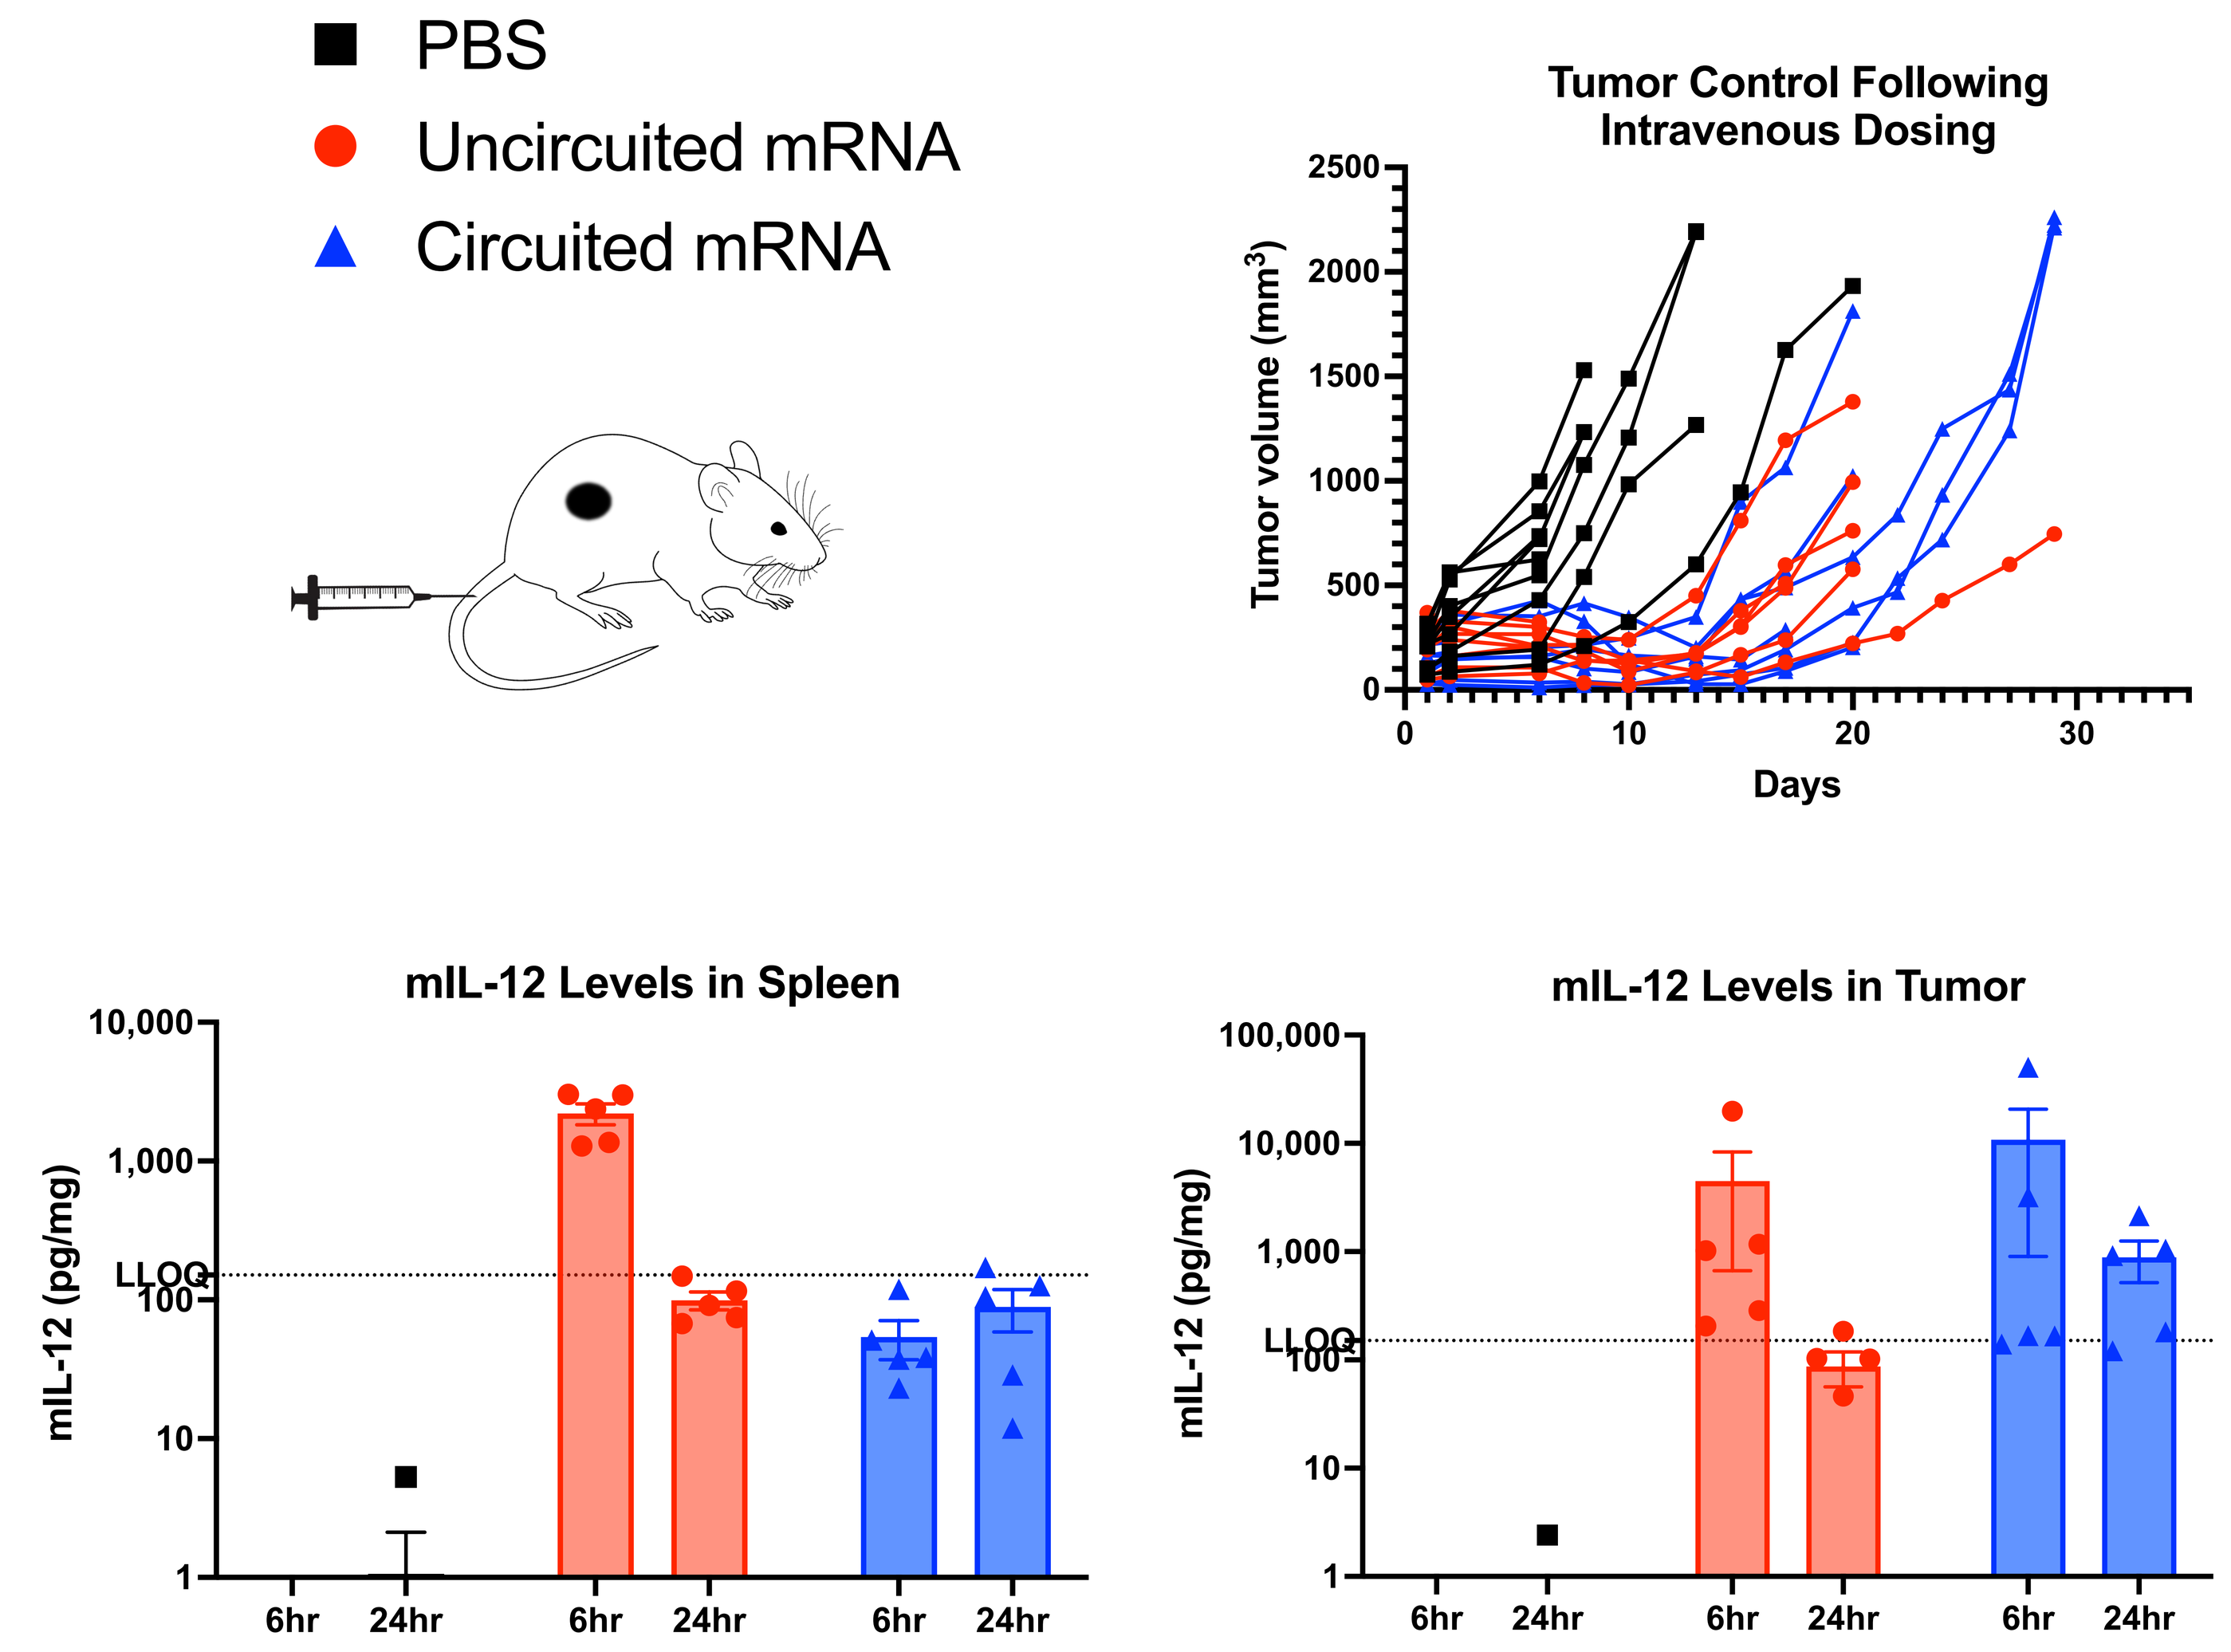
PBS
Uncircuited mRNA
Circuited mRNA
Tumor Control Following Intravenous Dosing
Tumor volume (mm³)
Days
mIL-12 Levels in Spleen
mIL-12 Levels in Tumor
mIL-12 (pg/mg)
LLOQ
6hr
24hr

- Mice were implanted subcutaneously with B16-F10 melanoma cells and were dosed intravenously with mRNA-LNP when tumors reached an average size of ~100-150 mm^3.
- Mice dosed with circuited mRNA significantly (≥ 40-fold) knocked down levels of mIL-12 in the spleen at Tmax, while mIL-12 levels in the tumor were comparable to the uncircuited cohort.
- The presence of the circuit did not impact efficacy, as tumor control was comparable with the uncircuited cohort.

CONCLUSIONS

- **STX-003 expresses its IL-12 payload in tumors while selectively repressing its expression in the spleen.**
- **STX-003 significantly reduces serum levels of IL-12 and IFN-γ in naïve mice and cynomolgus monkeys after intravenous dosing. In naïve mice, this correlates with improved tolerability.**
- **STX-003 demonstrates spleen-specific detargeting while maintaining effective tumor control upon systemic administration.**
- **These data demonstrate the potential of STX-003, a programmable mRNA circuit platform, for the development of precision mRNA therapies for cancer.**